D29

Integrated Efficacy and Safety Exposure Response (ER) Analysis of Tivozanib (Tivo) for the Treatment of Renal Cell Carcinoma (RCC)

Bradley Alexander McGregor,[1] Toni K. Choueiri,[1] Laurence Albiges,[2] Katy Beckermann,[3] Philippe Barthelemy,[4] Roberto Iacovelli,[5] Sheik Muhummud Fardeen Emambux,[6] Javier Molina-Cerrillo,[7] Benjamin Garmezy,[8] Pedro C. Barata,[9] Rana R. McKay,[10] Alex Chehrazi-Raffle,[11] Hans J. Hammers,[12] Daniel Yick Chin Heng,[13] Klaas Prins,[14] Bo Jin,[15] Monette Cotreau,[16] Edgar E. Braendle,[15] Claudia Lebedinsky,[15] Robert J. Motzer[17]

[1]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [2]Institut Gustave Roussy, Villejuif, France; [3]Vanderbilt University Medical Center, Nashville, TN, USA; [4]Institut de Cancerologie Strasbourg Europe, Strasbourg, France; [5]Università Cattolica del Sacro Cuore, Rome, Italy; [6]Centre Hospitalier Universitaire de Poitiers, Poitiers, France; [7]Hospital Universitario Ramon y Cajal, Madrid, Spain; [8]Sarah Cannon Research Institute, Nashville, TN, USA; [9]University Hospitals, Seidman Cancer Center, Cleveland, OH, USA; [10]Tulane University School of Medicine, New Orleans, LA, USA; [10]University of California, San Diego, San Diego, CA, USA; [11]City of Hope, Los Angeles, CA, USA; [12]University of Texas Southwestern, Dallas, TX, USA; [13]Tom Baker Cancer Centre, Calgary, AB, Canada; [14]qPharmetra, Nijmegen, the Netherlands; [15]AVEO Oncology, Boston, MA, USA; [16]MMC Biopartners, Rye, NH, USA; [17] Memorial Sloan Kettering Cancer Center, New York, NY, USA.

Introduction

- Tivo is a potent and highly selective oral vascular endothelial growth factor (VEGF) tyrosine kinase inhibitor (TKI) designed to optimize VEGF blockade and minimize off-target toxicities. Tivo is approved by the US Food and Drug Administration (FDA) for treatment of patients with relapsed/refractory RCC following ≥2 prior systemic therapies[1]
- The approved monotherapy starting dose is 1.34mg of Tivo (free base) once daily on days (D) 1-21 Q28D, with allowable dose modifications to manage adverse events. For patients with moderate hepatic impairment, starting dose is 0.89mg (free base) D1-21 Q28D
- Two phase 3 trials randomized patients with metastatic RCC 1:1 to receive 1.34 mg of tivo (free base*) vs sorafenib (in both trials)
 - **TIVO-1** (NCT01030783): patients received Tivo or sorafenib as initial targeted therapy[2]
 - **TIVO-3** (NCT02627963): patients received Tivo or sorafenib after progression on two or three systemic therapies[3]
- **TiNivo-2** (NCT04987203) randomized patients with advanced RCC 1:1 to Tivo 0.89 mg D1-21 q28D plus nivolumab (Nivo) 480 mg q4 weeks, or Tivo (1.34 mg D1-21 q28D)[4]
 - Reduced dose of tivo in combination arm was agreed with regulatory authorities due to potential risk of higher rate of grade 3/4 hypertension[4]

Study Objective

- The current analyses were thus undertaken to better understand the relationship between Tivo exposure with PFS, tumor size (TS), and hypertension (HTN; safety endpoint) using Population pharmacokinetic (PopPK) and exposure response (ER) modeling in mRCC

*TIVO-1 and TIVO-3 patient received 1.5mg of tivozanib hydrochloride (equivalent to 1.34 mg of tivozanib free base)

Methods

We present exploratory analyses evaluating the PK of Tivo in a dataset that includes TiNivo-2, TIVO-1, and TIVO-3, in patients that received Tivo at 1.34mg and 0.89mg. Patient baseline characteristics from these studies are summarized in **Table 1**.

- PopPK and ER models were developed with data from Tivo-1[2], and Tivo-3[3] (in-house data)
- TiNivo-2 trial results were integrated to update the PopPK and ER models for PFS (Cox proportional hazard), TS (sum of longest diameters longitudinal model), and HTN (logistic regression) and to simulate the ER-based risk/benefit profile of Tivo

PFS: Cox Proportional Hazard Model

The effect of Tivo exposure on PFS was modeled in 776 patients from TIVO-1, TIVO-3, and TiNivo-2

- Tivo exposure was represented by model-predicted average Tivo serum concentration (C_{avg}) in patients in Cycle 2
- Exposure ranges were grouped into quartiles based on equal numbers of samples resulting in C_{avg} ranges: 13.9 to 38.4, 38.4 to 47.9, 47.9 to 62.0, and 62.0 to 177 ng/mL

TS: sum of longest diameters longitudinal model

Changes in TS (by radiographic measurement) and ER were investigated for patients treated with Tivo mono- and Tivo-Nivo combination therapies by plotting baseline and post-treatment TS measurements for each exposure quartile (total of 722 patients)

HTN: Logistic Regression Model

Associations between Tivo exposure and any grade HTN were investigated for Tivo mono- and Tivo-Nivo combination treated patients (total of 814 patients from phase 1 to 3 trials)

Results

Table 1. Baseline Patient Demographics of TIVO-1, TIVO-3, and TiNivo-2[2,3,4]

	TIVO-1 (N=517)		TIVO-3 (N=350)		TiNivo-2 (N=343)	
Characteristic	Tivo (1.34 mg*) (n=260)	Sorafenib (n=257)	Tivo (1.34 mg*) (n=175)	Sorafenib N=175	Tivo (1.34 mg) (n=172)	Tivo (0.89 mg**)-NIVO (n=171)
Median age, yrs (range)	59 (23–83)	59 (23–85)	62 (34–88)	63 (30–90)	63 (33–82)	64 (37–87)
Male, n (%)	185 (71)	189 (74)	126 (72)	128 (73)	134 (78)	125 (73)
ECOG PS 0-1, n (%)	230 (100)	257 (100)	173 (100)	168 (98)[a]	172 (100)	170 (100)[b]
Race, n (%)						
White	249 (96)	249 (97)	165 (94)	167 (95)	107 (62)	112 (65)
Asian	10 (4)	8 (3)	10 (6)	8 (5)	0	1 (<1)
Black	1 (<1)	0			8 (5)	2 (1)
Not reported	-	-			57 (33)	56 (33)

*TIVO-1 and TIVO-3 patient received 1.5mg of tivozanib hydrochloride (equivalent to 1.34 mg of tivozanib free base; **TiNivo-2 patients received 0.89mg of tivozanib hydrochloride (equivalent to 1.0mg of tivozanib free base); ECOG PS, Eastern Cooperative Oncology Group Performance Status; [a]Two patients had ECOG PS 2; [b] One patient missing.

Tivozanib Exposure Response

- The range of observed Tivo exposures (C_{avg}) was represented by the quartiles, each with the same number of patients
- A strong correlation between higher concentrations of Tivo and duration of PFS was observed (**Figure 1**)
 - Median (m) PFS of 9.7 months was observed in patients with the highest C_{avg}
 - mPFS of 5.6 months in patients with the lowest C_{avg}
- A correlation between Tivo C_{avg} and change in TS from baseline was also observed (**Figure 2**)
 - Patients in the TiNivo-2 combination therapy arm who received 0.89mg Tivo are mostly clustered in the lowest C_{avg} quartile
 - As C_{avg} quartiles of Tivo increase, patients' TS decreases, indicating higher Tivo results in an increased treatment response
 - Blue line is the Loess smooth fit to the data
- PopPK modelling shows that Nivo does not affect Tivo PK (**Figure 3**)
- Incidence of any grade or grade 3 hypertension was not significantly different for patients treated with 1.34mg or 0.89mg of Tivo (**Figure 4**)

Strong exposure response relationship between cycle 2 Tivozanib serum C_{avg} and PFS

Figure 1. PFS stratified by Tivo Cycle 2 C_{avg} quartile

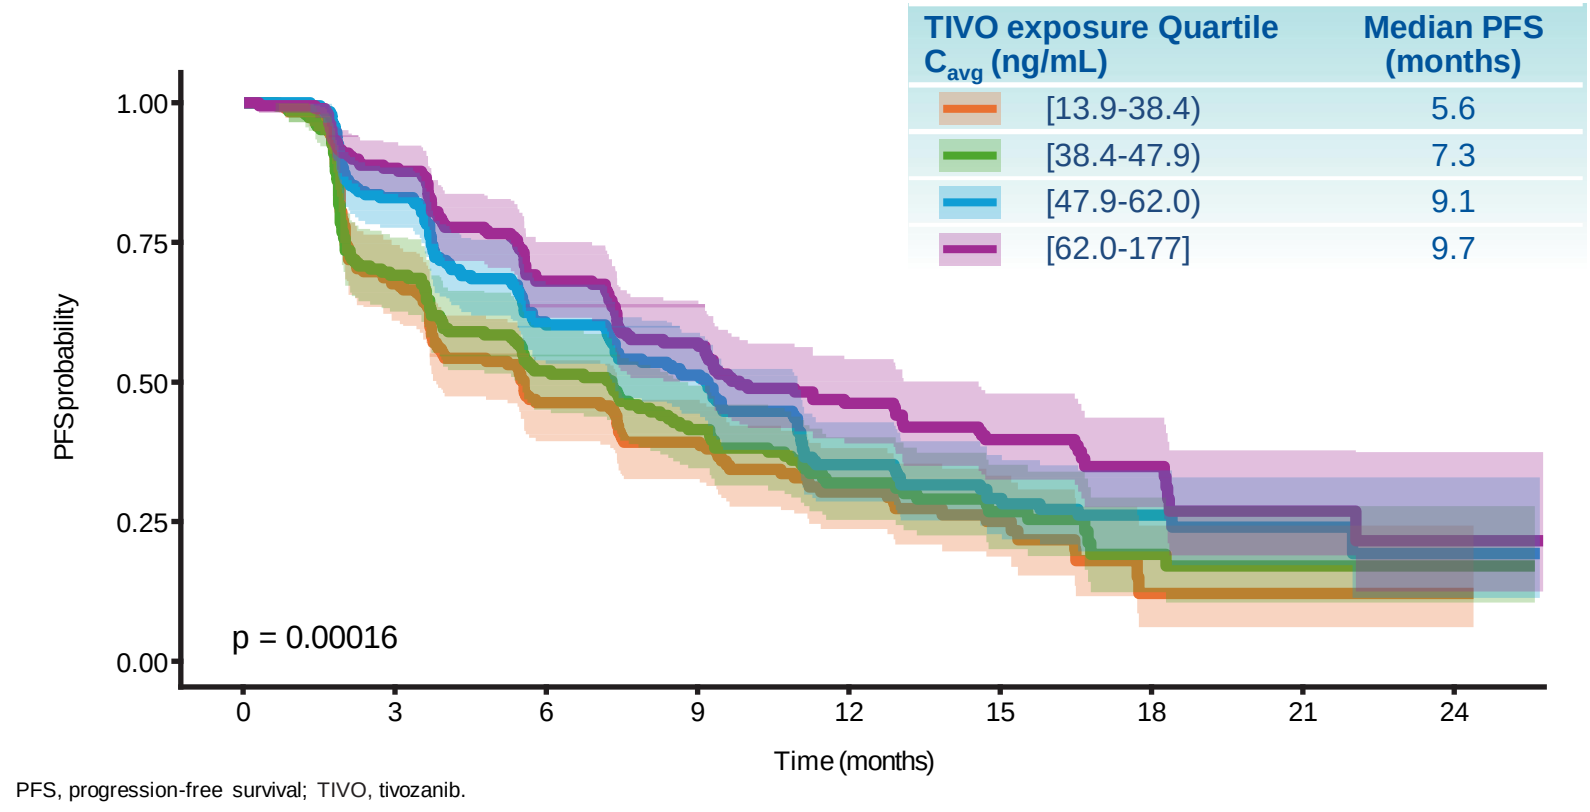
PFSprobability
1.00
0.75
0.50
0.25
0.00
p = 0.00016
0
3
6
9
12
15
18
21
24
Time (months)

TIVO exposure Quartile C_{avg} (ng/mL)	Median PFS (months)
[13.9-38.4)	5.6
[38.4-47.9)	7.3
[47.9-62.0)	9.1
[62.0-177]	9.7

PFS, progression-free survival; TIVO, tivozanib.

Tivozanib PK is unaltered by the addition of Nivolumab

Figure 3. Observed Tivo serum concentrations and the modeling fit

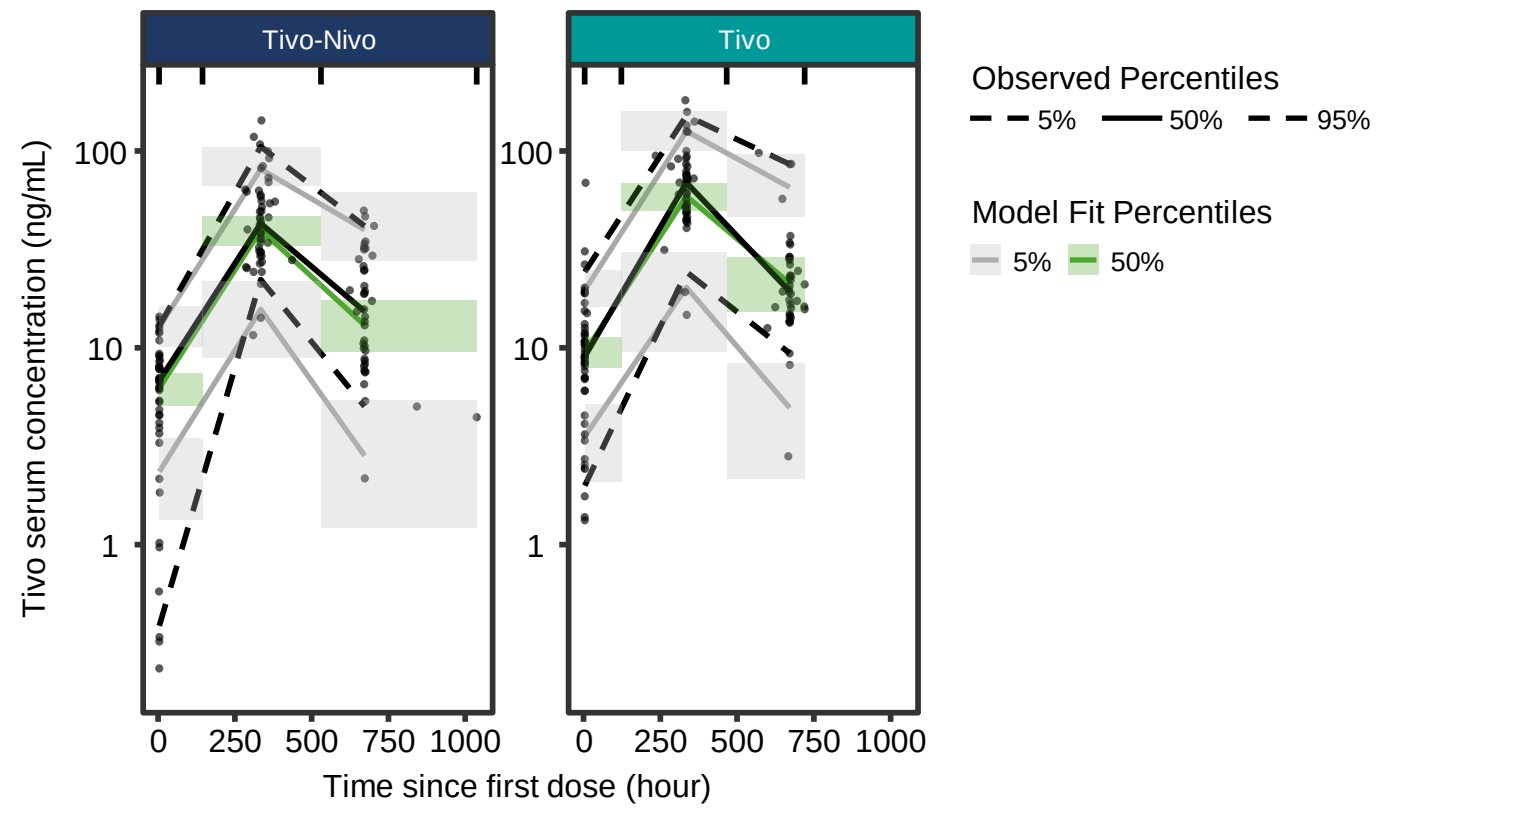
Tivo-Nivo
Tivo
Tivo serum concentration (ng/mL)
100
10
1
0 250 500 750 1000
Time since first dose (hour)
Observed Percentiles
5%
50%
95%
Model Fit Percentiles
5%
50%

Higher Tivozanib exposure (C_{avg}), greater tumor response

Figure 2. Tivo C_{avg} quartile correlations with changes in TS and best overall response (BOR) in TS

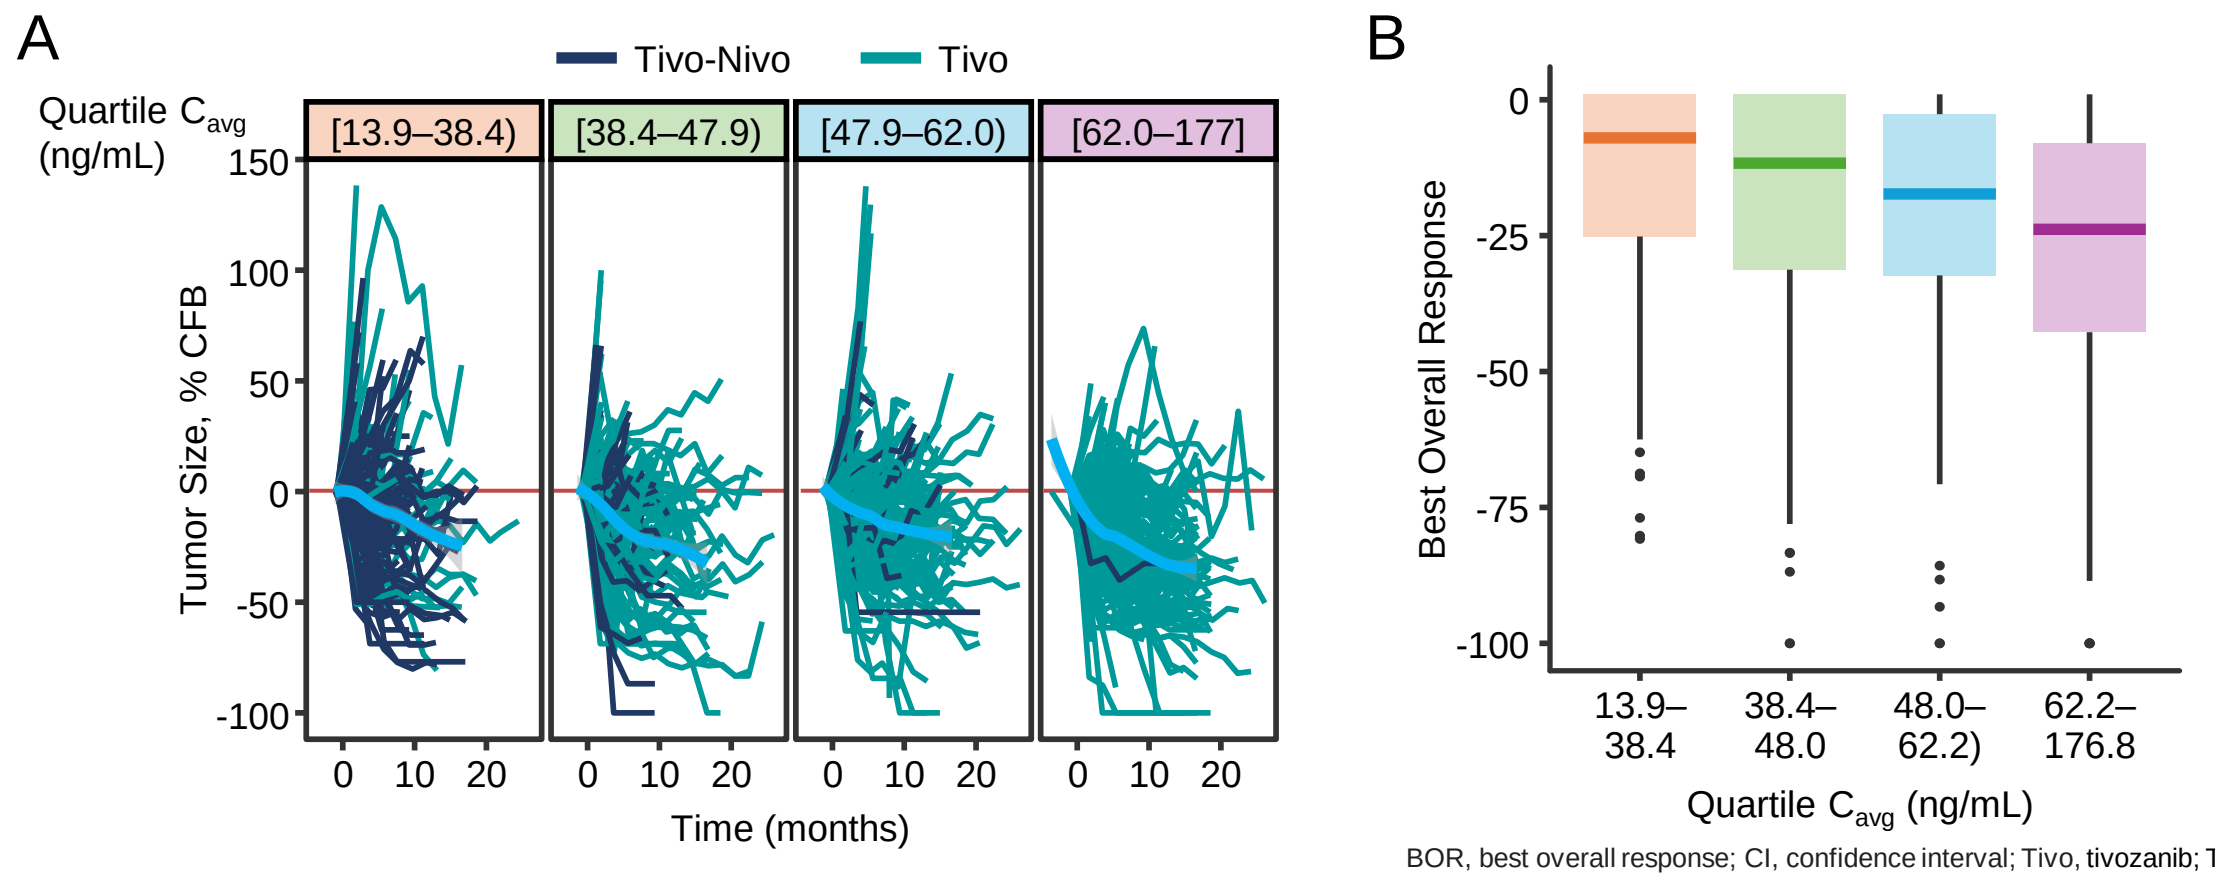
A
Tivo-Nivo
Tivo
Quartile C_{avg} (ng/mL)
[13.9–38.4)
[38.4–47.9)
[47.9–62.0)
[62.0–177]
Tumor Size, % CFB
150
100
50
0
-50
-100
0 10 20
Time (months)
B
Best Overall Response
0
-25
-50
-75
-100
13.9–38.4
38.4–48.0
48.0–62.2)
62.2–176.8
Quartile C_{avg} (ng/mL)

Quartile C_{avg} (ng/mL)	N	Median BOR (95% CI)
[13.9–38.4)	176	-7.02 (1.03– -16)
[38.4–47.9)	173	-11.7 (-2.05– -21.3)
[47.9–62.0)	185	-17.3 (-8.27– -26.4)
[62.0–177]	183	-23.8 (-14.1– -33.5)

BOR, best overall response; CI, confidence interval; Tivo, tivozanib; TS, tumor size.

ER modeling analysis of hypertension

Figure 4.

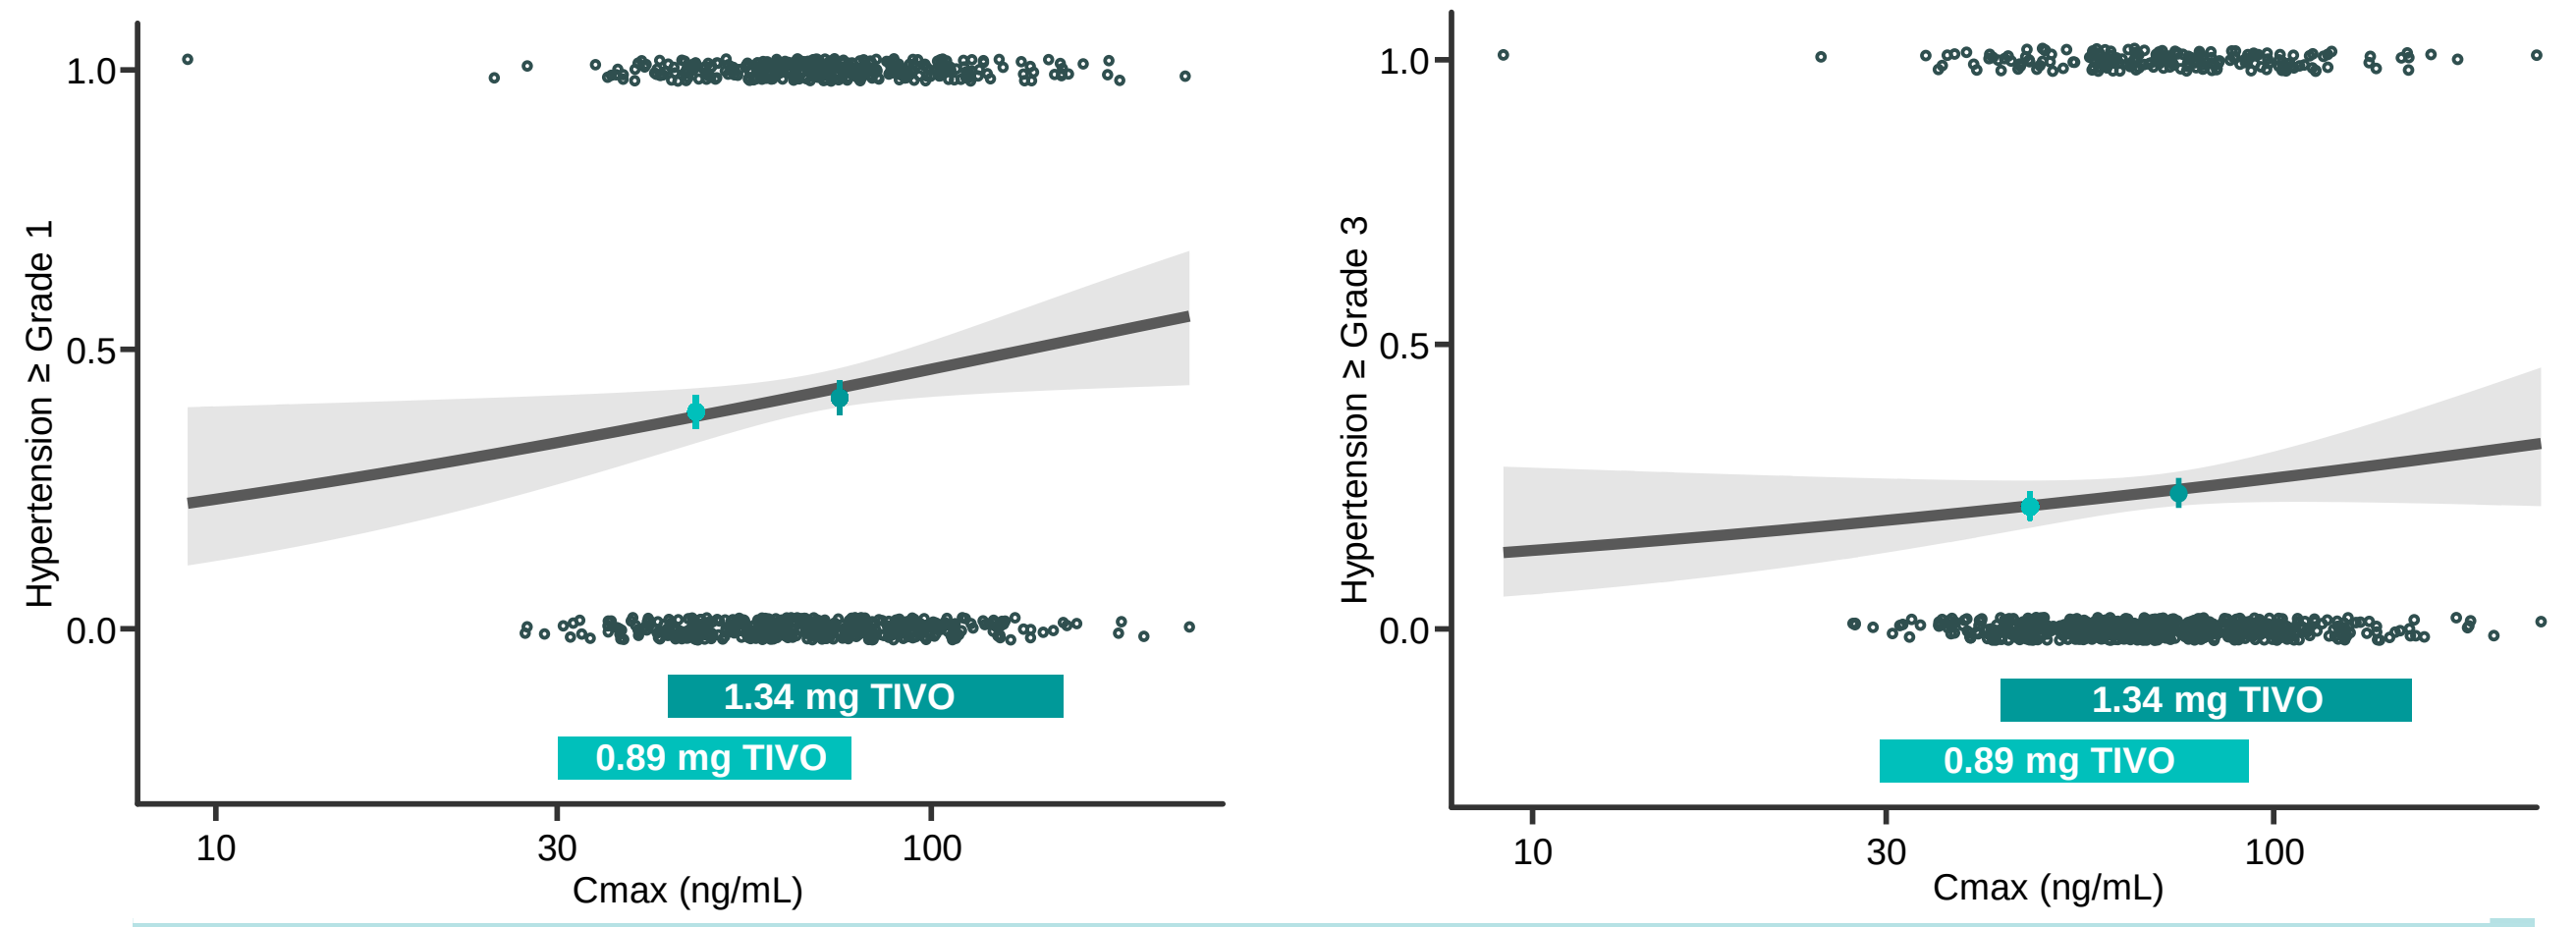
Hypertension ≥ Grade 1
Hypertension ≥ Grade 3
1.0
0.5
0.0
1.34 mg TIVO
0.89 mg TIVO
10
30
100
Cmax (ng/mL)

	Hypertension (any grade)		Hypertension (Grade ≥3)	
Tivo	Incidence, % (95% PI)	C_{max} (ng/mL)	Incidence, % (95% PI)	C_{max} (ng/mL)
1.34 mg	0.413 (0.383–0.444)	74.4 (42.8–153)	0.238 (0.213–0.265)	74.4 (42.8–153.0)
0.89 mg	0.388 (0.358–0.419)	46.9 (31.6–76.5)	0.215 (0.191–0.242)	46.9 (31.6–76.5)

Logistic regression model using 1000 resampled Tivo exposures for each treatment arm; Tivo, tivozanib.

Conclusions

- ER models shows that Tivo 1.34mg provides greater anti-tumor activity than 0.89mg. The hypertension incidence seemed to be comparable.
- These ER data suggest that a dose of 1.34mg Tivo provides a greater decrease in TS and may be more tolerable.
- The results from the TiNivo-2 data set further confirmed that re-challenge with immunotherapy does not add benefit and optimal dosing of Tivo provided the highest clinical benefit following progression on immunotherapy.

References

1. FOTIVDA® (tivozanib) Prescribing Information. 2024.
2. Motzer RJ, *et al. J Clin Oncol.* 2013; 31:3791–3799.
3. Rini BI, *et al. Lancet.* 2020; 21:95–104
4. Choueiri TK, *et al. Lancet.* 2024; 404:1309–1320.

Acknowledgments

We express our gratitude to the patients and their caregivers for their invaluable contributions. We also extend our thanks to the clinical study teams, investigators, and steering committee. This study is sponsored by AVEO Oncology. Nivolumab was provided by Bristol Myers Squibb. Editorial assistance was provided by Amber Wood, of Nucleus Global, an Inizio Company, and funded by AVEO Oncology.

Presented at the American Society of Clinical Oncology Genitourinary Cancers Symposium; February 13-15, 2025; San Francisco, CA

E-mail: Bradley_McGregor@DFCI.HARVARD.EDU