Phase 2 clinical evaluation of preclinically defined biomarkers for vascular endothelial growth factor tyrosine kinase inhibitor tivozanib in renal cell carcinoma

Thomas E. Hutson,[1] W. Kimryn Rathmell,[2] Bin Feng,[3] Murray O. Robinson,[3] Jeno Gyuris,[3] Jie Lin,[3] Toni K. Choueiri[4]

[1]Texas Oncology Baylor Charles A. Sammons Cancer Center, Dallas, TX, USA; [2]University of North Carolina at Chapel Hill, Chapel Hill, NC, USA; [3]AVEO Oncology, Cambridge, MA, USA; [4]Dana-Farber Cancer Institute, Boston, MA, USA

Poster No: 233P

Background

- Despite significant effort, identifying predictive biomarkers for vascular endothelial growth factor (VEGF)-targeted therapies remains a challenge[1]
- Utilizing population-based tumor models, we identified and characterized a specific population of infiltrating myeloid cells associated with intrinsic resistance to tivozanib[2]
 - Tivozanib is a potent, investigational, selective tyrosine kinase inhibitor of VEGF receptor-1, -2, and -3 that was evaluated for the treatment of advanced renal cell carcinoma (RCC)[3,4]
- RNA and protein myeloid cell biomarkers derived from preclinical studies were evaluated in a Phase 2 single-arm trial of tivozanib monotherapy in nephrectomized, targeted-therapy-naïve RCC patients (AV-951-10-202/NCT01297244)

Study Objective

- One of the objectives of the study was to evaluate a myeloid-associated resistance hypothesis in human tivozanib-treated clear cell RCC (ccRCC) using both RNA and immunohistochemistry (IHC)-based markers

Methods

- Tivozanib efficacy studies of individual tumors from a molecularly characterized population-based model[2] found intrinsic resistance in 16/25 (64%) of the tumors
 - Analysis of top pathways associated with a 200-gene profile (**Figure 1**) revealed the presence of an infiltrating myeloid population associated with resistance

Figure 1. Tivozanib Resistance Signature.

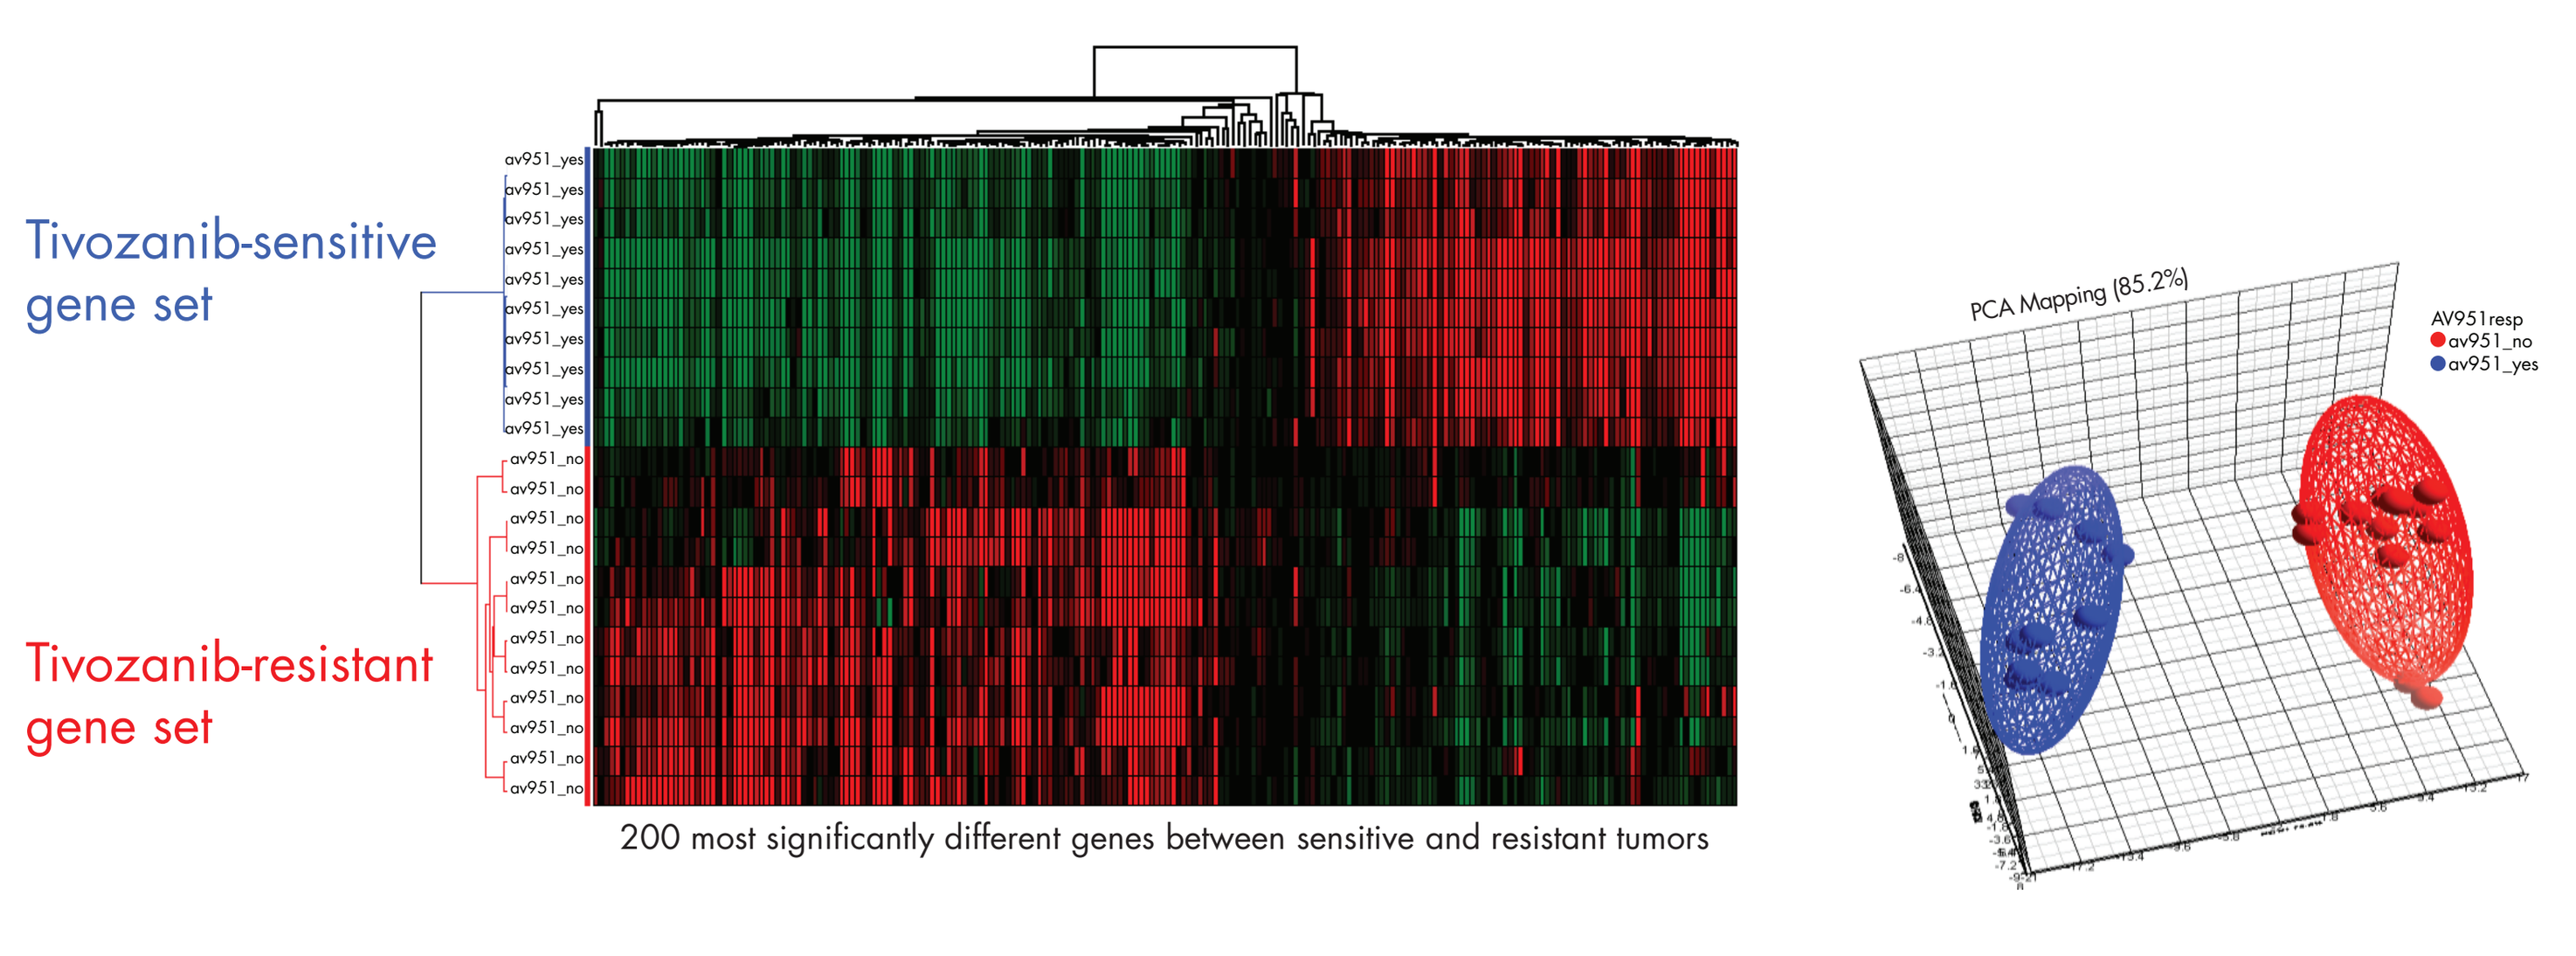
Tivozanib-sensitive gene set
Tivozanib-resistant gene set
200 most significantly different genes between sensitive and resistant tumors
PCA Mapping (85.2%)
AV951resp
av951_no
av951_yes

- Expression-based correlation analyses of myeloid genes in expression data from 600 human tumors resulted in a 42-gene myeloid signature (**Figure 2A**); 24 genes were formatted for Taqman PCR quantitation in human formalin-fixed paraffin-embedded (FFPE) sections
- Patients enrolled in AV-951-10-202/NCT01297244 received 1.5-mg tivozanib orally, once daily (QD) for 3 weeks, followed by 1 week off-treatment for 28-day cycles[5] and prespecified biomarkers were evaluated
- Successfully obtained samples were assayed by qRT-PCR (Asuragen, Inc., Austin, TX); the aggregate score for the 24 genes was determined for each patient (**Figure 2B**)
 - Median signature score was used as the cut off for the biomarker
- CD68 positive-infiltrating myeloid cells were quantified by IHC (Aperio Scanscope, Leica Microsystems Inc., Buffalo Grove, IL)
 - Median was used as the cut off for the biomarker
- RNA signatures were evaluated for prognostic impact in a dataset collected prior to the use of VEGF-targeted therapies[6]

Figure 2A. Forty-two–Gene Myeloid Signature is Coherent in All Human Tumors and Suggests Broad Presence of Resistance-Associated Myeloid Infiltration.[a]

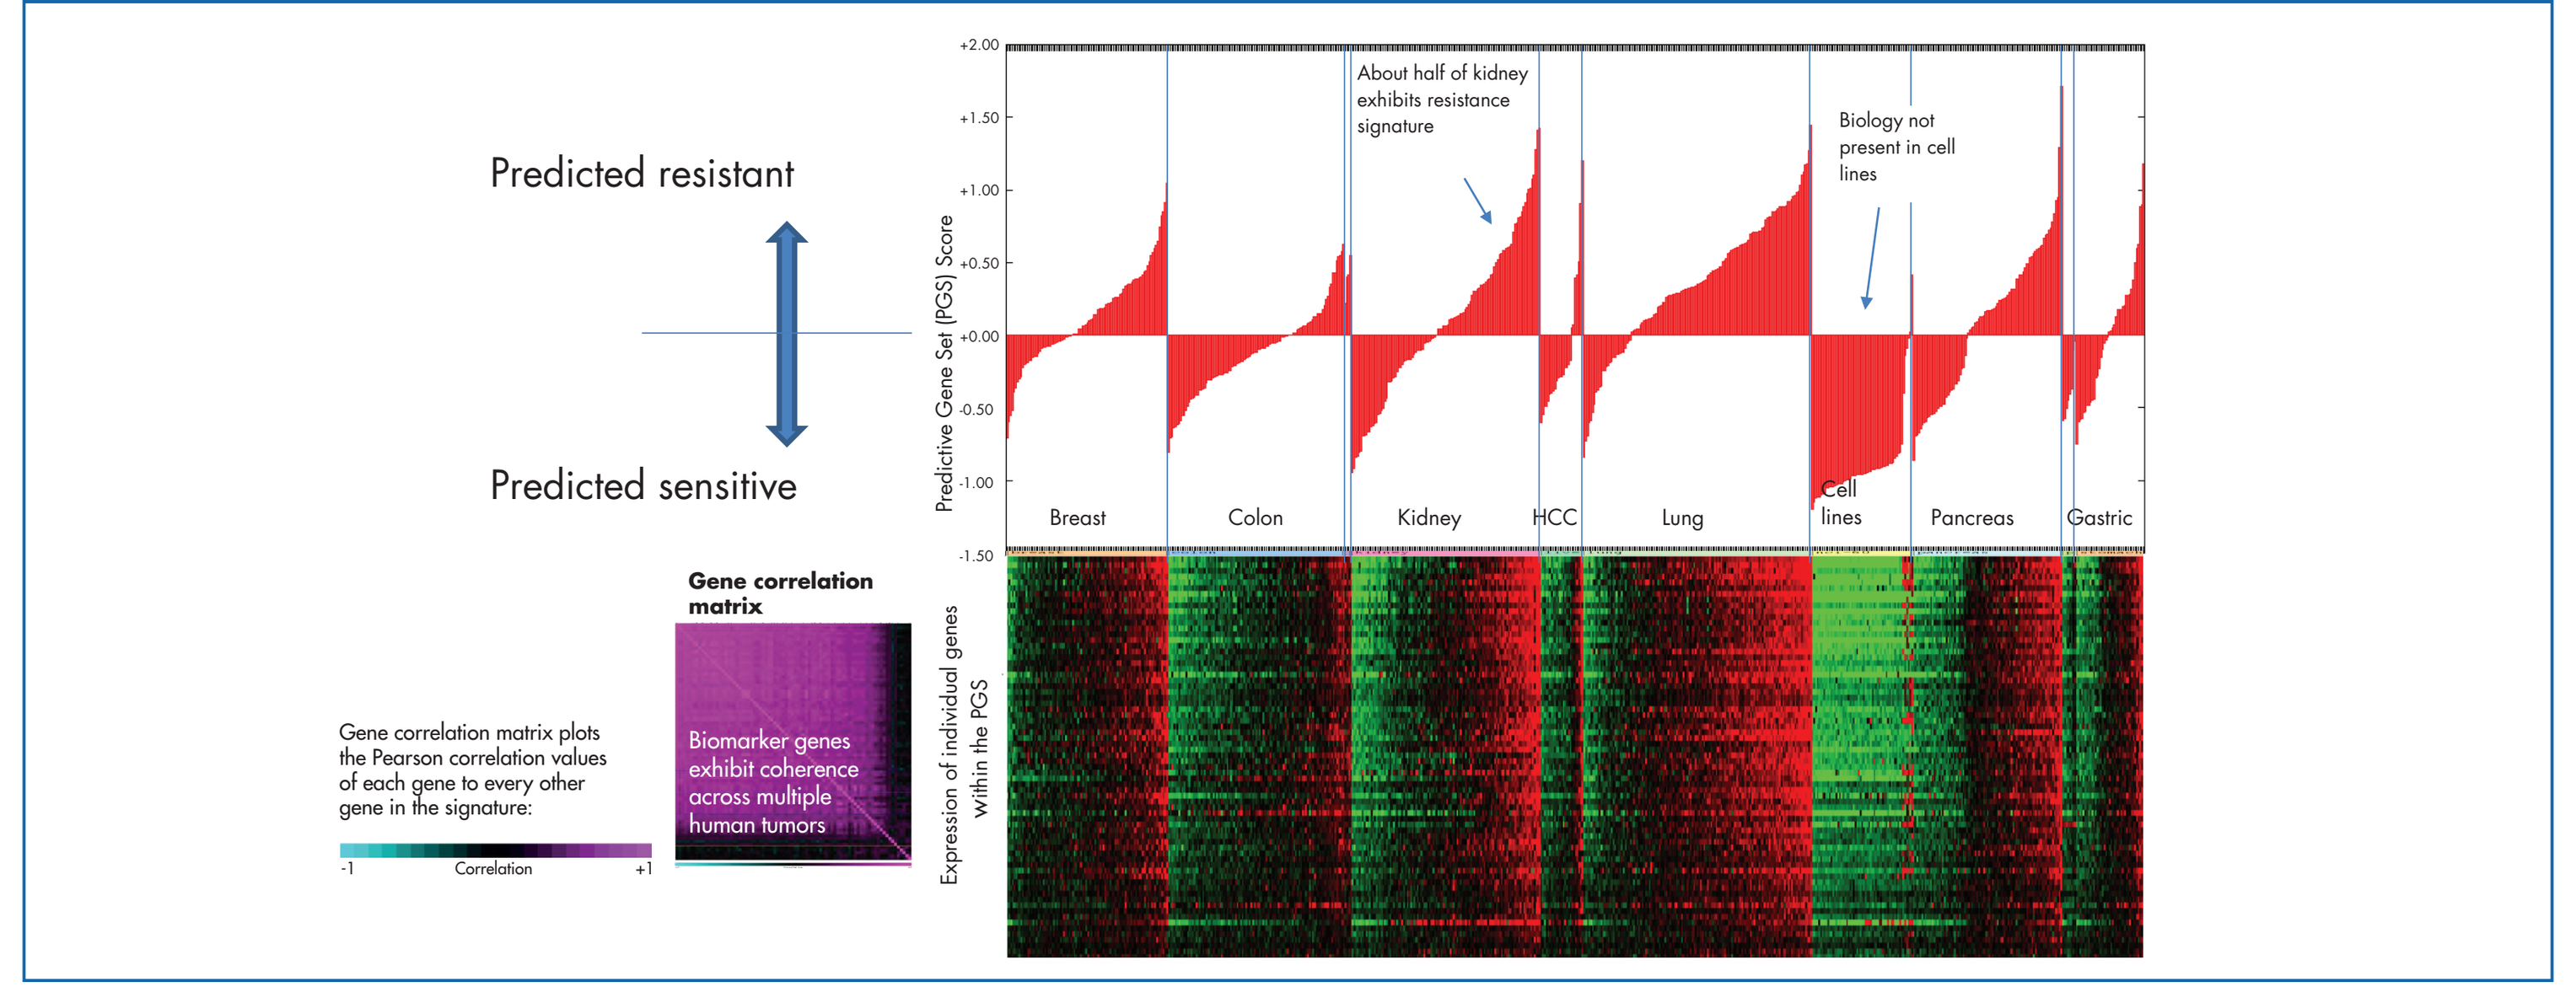
Predicted resistant
Predicted sensitive
Predictive Gene Set (PGS) Score
About half of kidney exhibits resistance signature
Biology not present in cell lines
Breast
Colon
Kidney
HCC
Lung
Cell lines
Pancreas
Gastric
Gene correlation matrix
Gene correlation matrix plots the Pearson correlation values of each gene to every other gene in the signature:
Correlation
Biomarker genes exhibit coherence across multiple human tumors
Expression of individual genes within the PGS

[a]Predictive gene set score was calculated by averaging expression values of each individual gene.

Figure 2B. Taqman PCR Data and Signature Score for Each of the Indicated Genes in Each Patient.

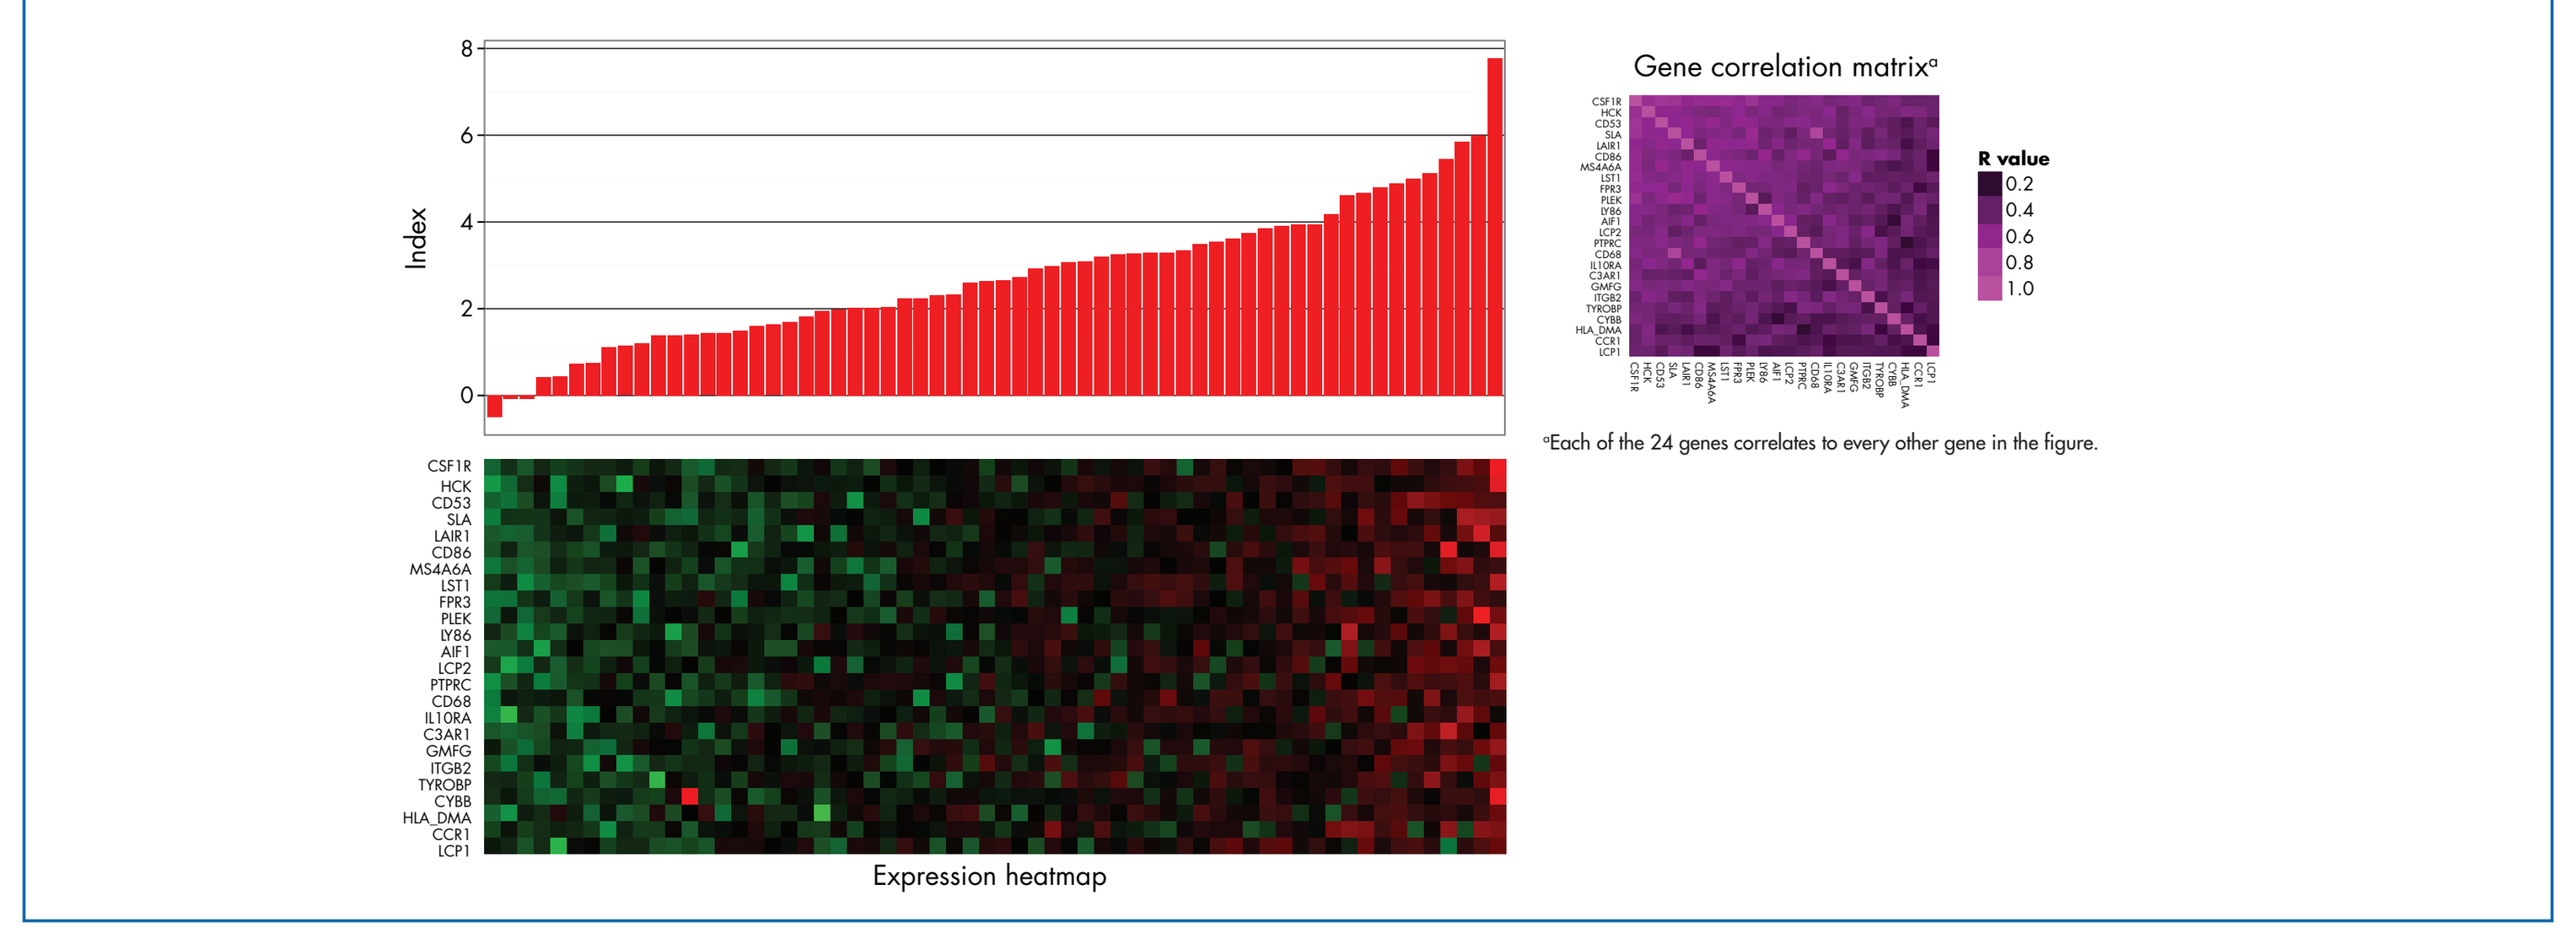
Index
Expression heatmap
Gene correlation matrix[a]
R value
[a]Each of the 24 genes correlates to every other gene in the figure.

Results

- One hundred five patients were enrolled, of which 90 patients had clear cell (cc) histology
 - Intent-to-treat (ITT) and ccRCC progression-free survival (PFS) were both 9.7 months (**Figure 3**)
 - ccRCC samples that passed quality check (RNA, 63; IHC, 66) were analyzed (**Figure 4**)

Figure 3. Kaplan-Meier Survival Plots: ITT and ccRCC Populations.

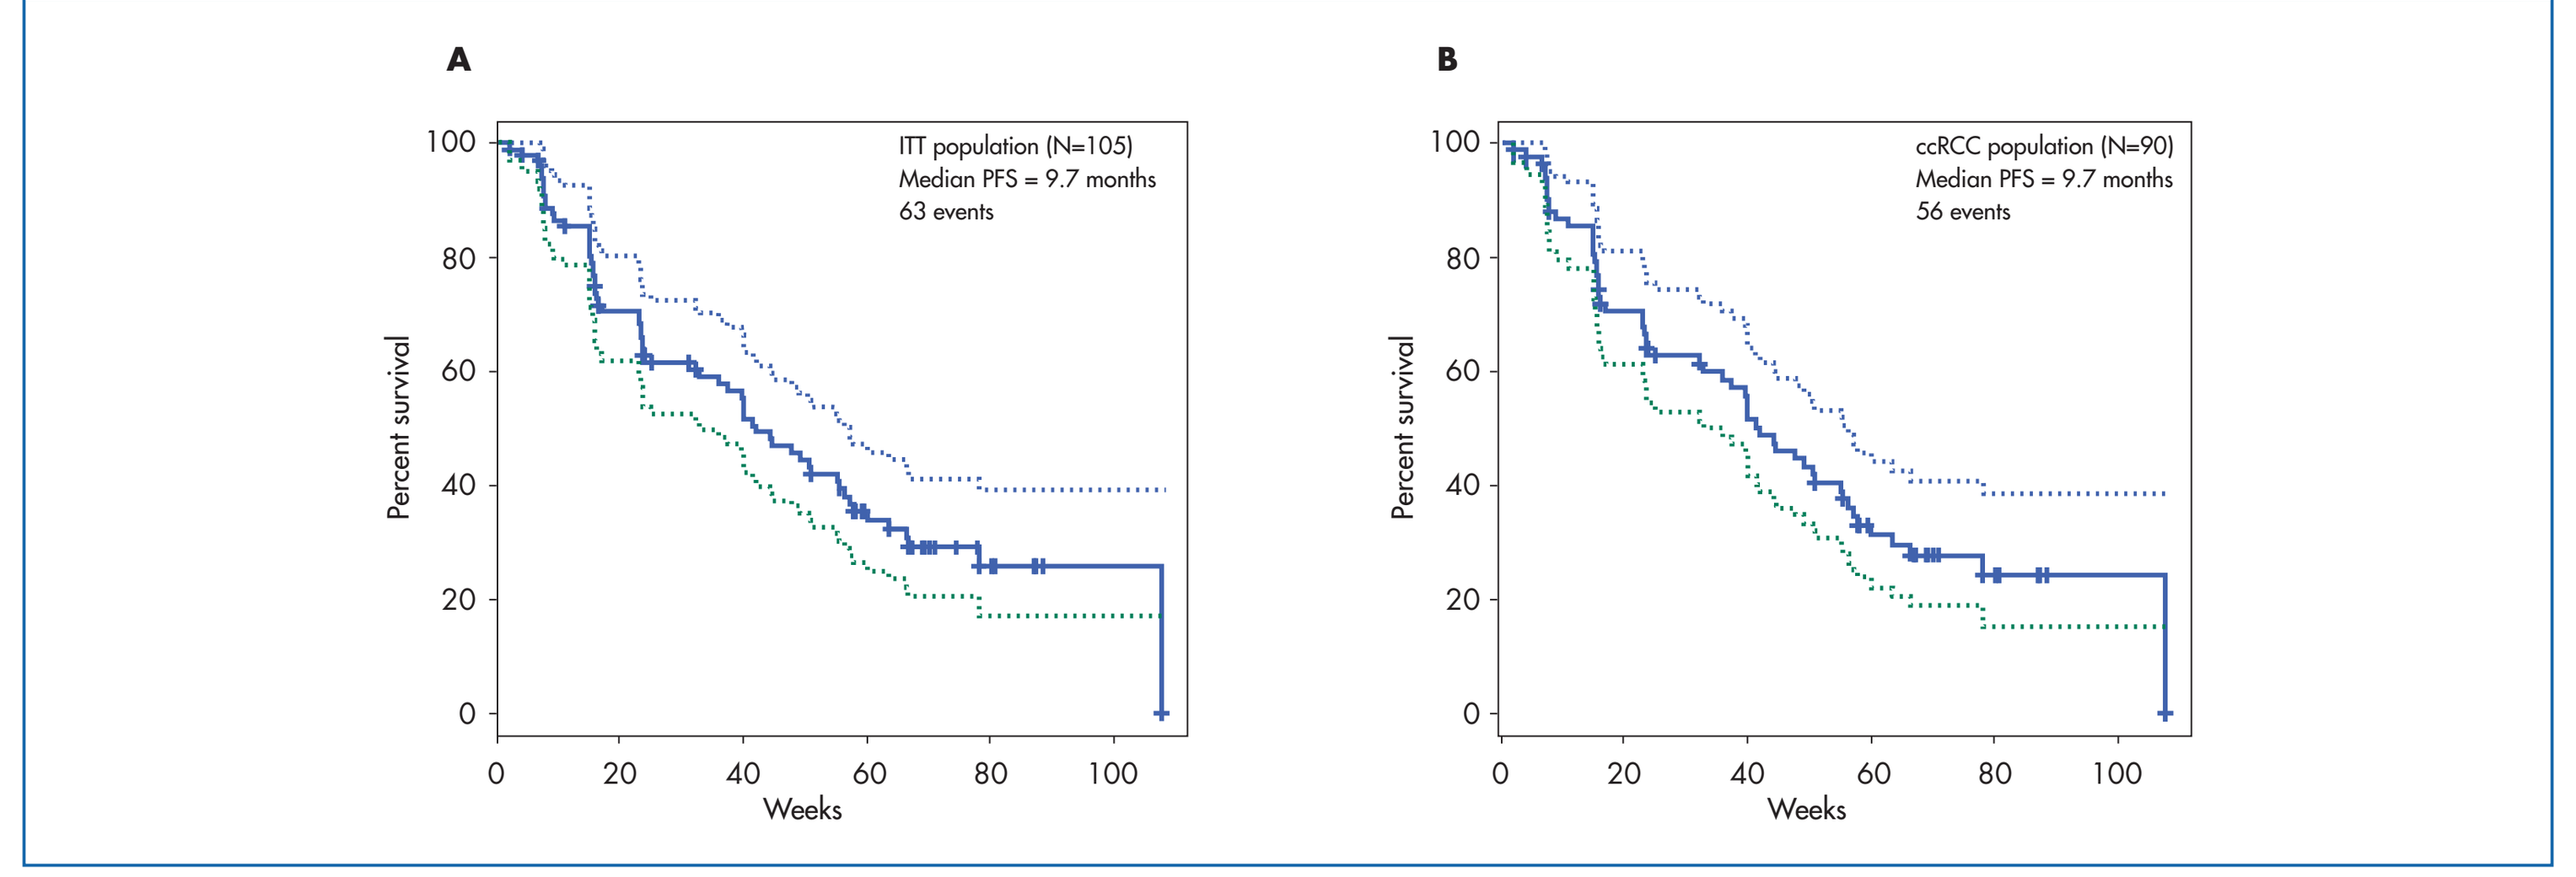
A
ITT population (N=105)
Median PFS = 9.7 months
63 events
B
ccRCC population (N=90)
Median PFS = 9.7 months
56 events
Percent survival
Weeks

ccRCC, clear cell renal cell carcinoma; ITT, intent-to-treat; PFS, progression-free survival.

Figure 4. Patient Samples Analyzed.

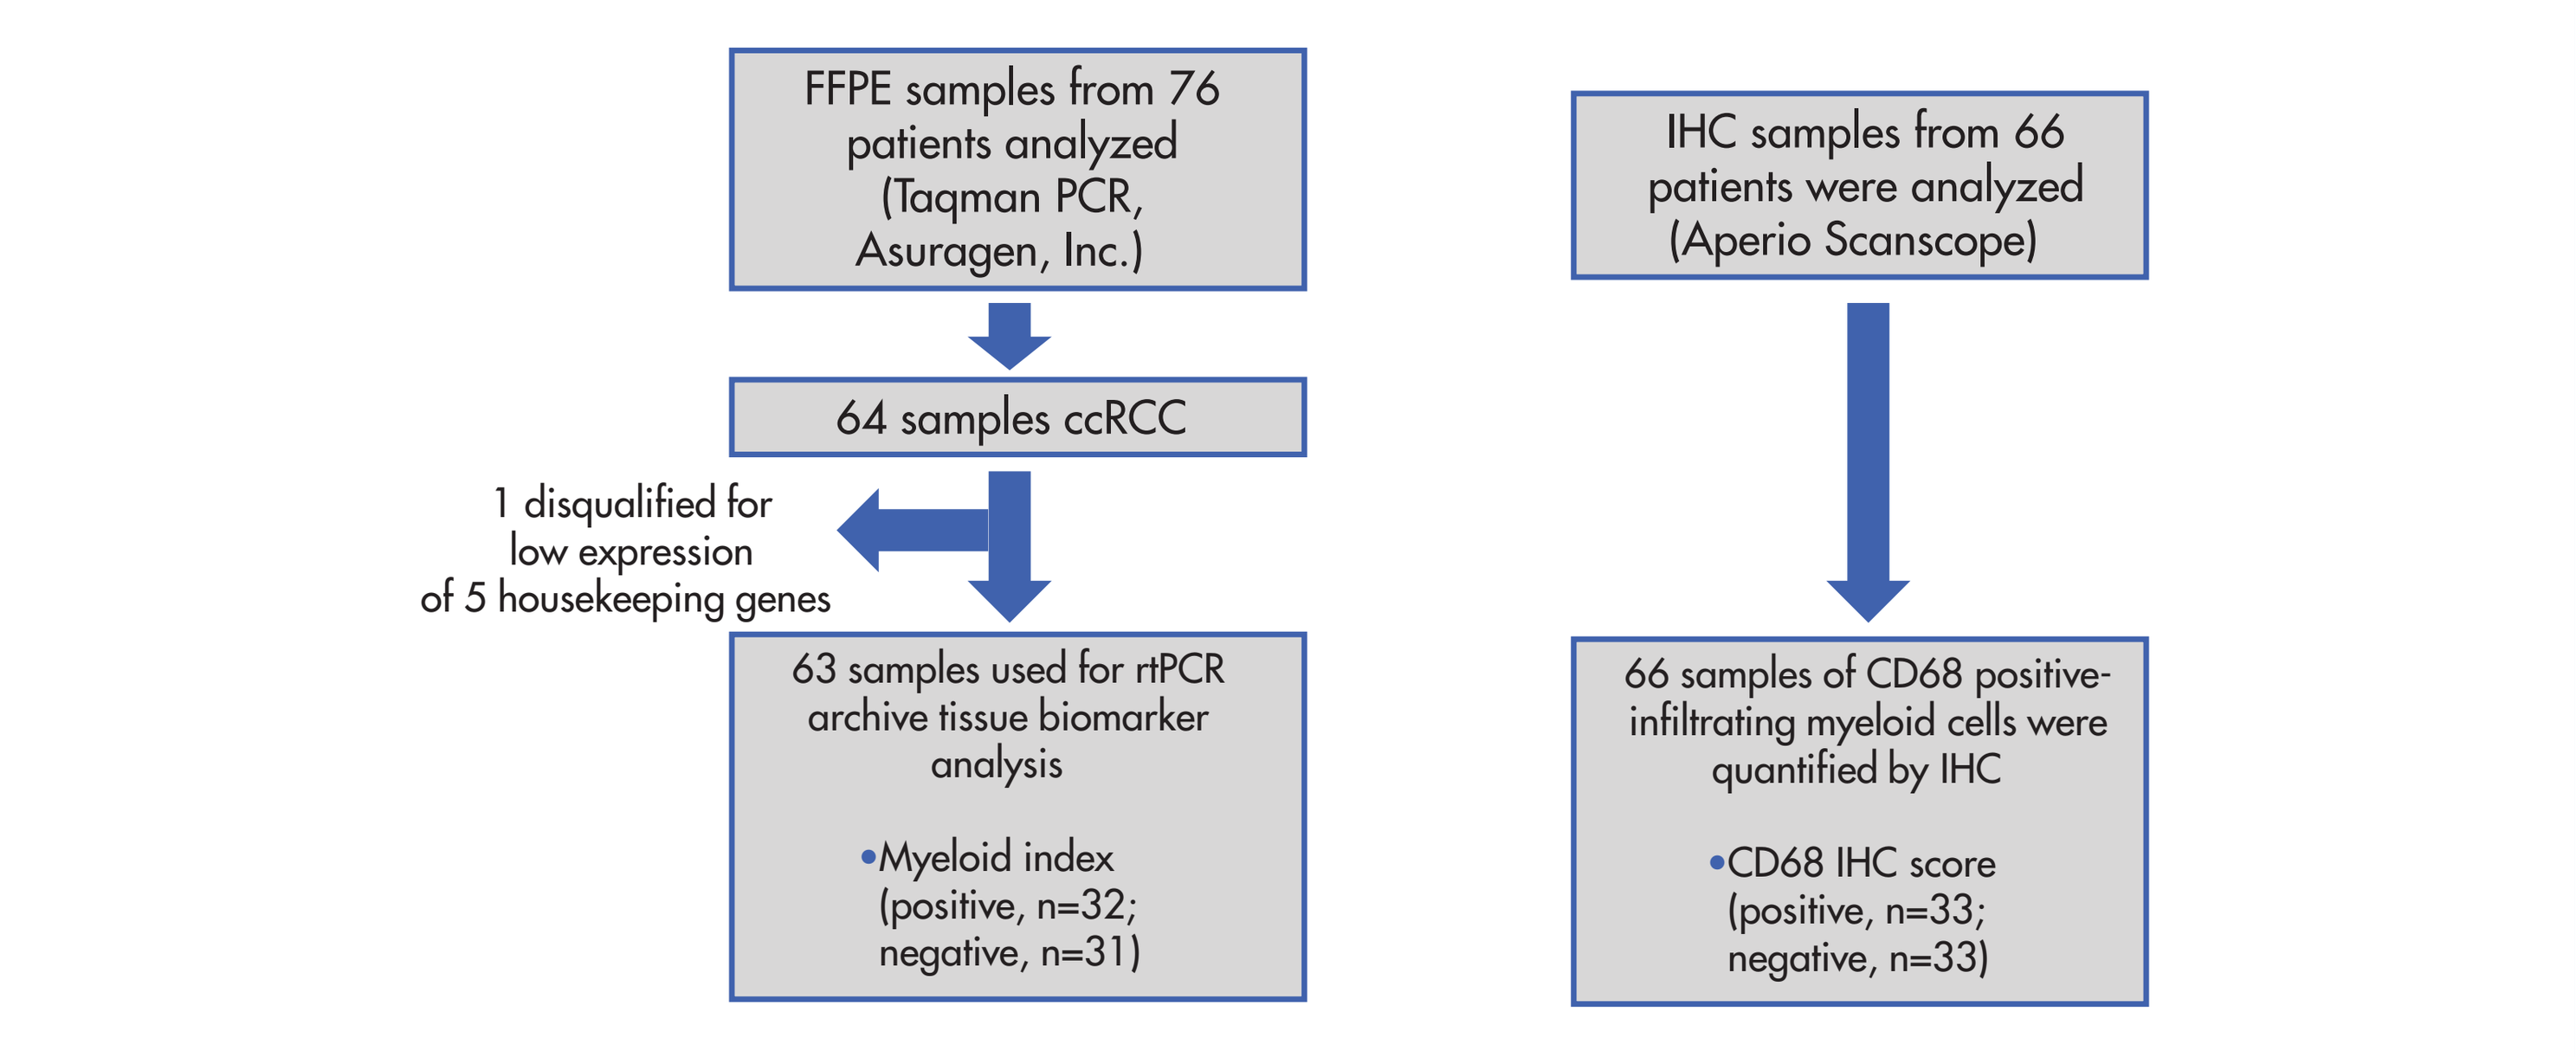
FFPE samples from 76 patients analyzed (Taqman PCR, Asuragen, Inc.)
64 samples ccRCC
1 disqualified for low expression of 5 housekeeping genes
63 samples used for rtPCR archive tissue biomarker analysis
•Myeloid index (positive, n=32; negative, n=31)
IHC samples from 66 patients were analyzed (Aperio Scanscope)
66 samples of CD68 positive-infiltrating myeloid cells were quantified by IHC
•CD68 IHC score (positive, n=33; negative, n=33)

ccRCC, clear cell renal cell carcinoma; FFPE, formalin-fixed paraffin-embedded; IHC, immunohistochemistry; PCR, polymerase chain reaction; rtPCR, reverse transcriptase polymerase chain reaction.

Myeloid Index

- Low myeloid index is associated with significantly longer PFS (**Figure 5**)
 - Longer PFS based on median cutoff (PFS 14.7 vs 8.3 months, hazard ratio [HR] 0.49, *P*=0.035; 95% CI 0.25–0.96) and as a continuous variable (*P*=0.03; N=63)
 - The gene signature did not exhibit a prognostic effect in a historical dataset[6]

Figure 5. Kaplan-Meier Survival Plot, Based on Myeloid Index.

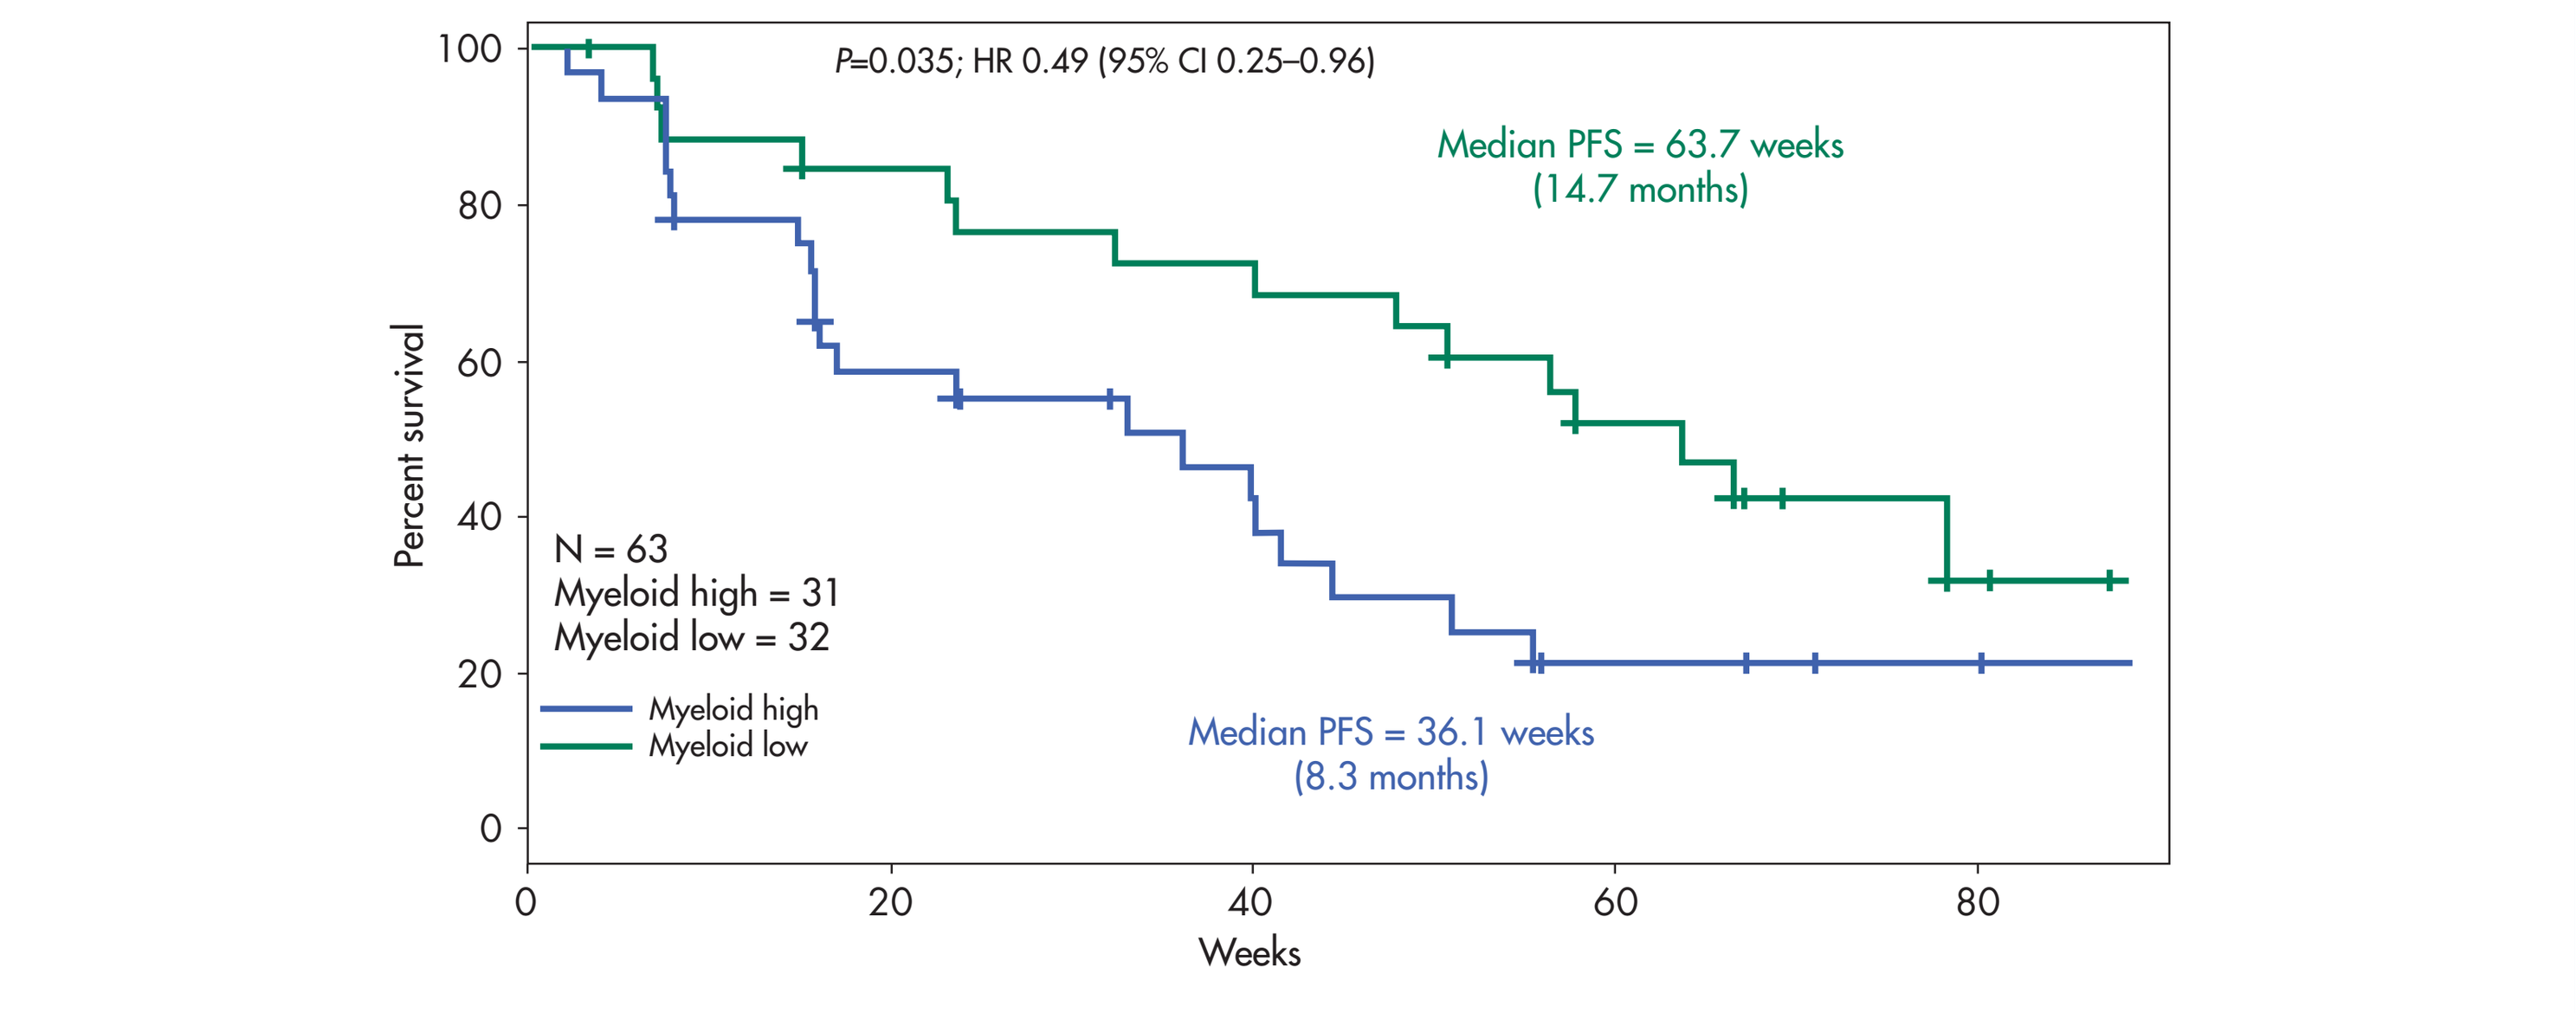
P=0.035; HR 0.49 (95% CI 0.25–0.96)
Median PFS = 63.7 weeks (14.7 months)
Median PFS = 36.1 weeks (8.3 months)
N = 63
Myeloid high = 31
Myeloid low = 32
Myeloid high
Myeloid low
Percent survival
Weeks

PFS, progression-free survival.

CD68 Score

- The CD68 IHC score exhibited a similar trend but was not significant (**Figure 6**)
 - Median cutoff PFS was 13.3 versus 9.2 months (HR 0.55, *P*=0.067; 95% CI 0.28–1.05; continuous *P*=0.057, N=66)
 - Myeloid low is represented as biomarker positive as seen in example IHC micrographs (**Figure 7**)

Figure 6. Kaplan-Meier Survival Plot, Based on CD68 IHC.

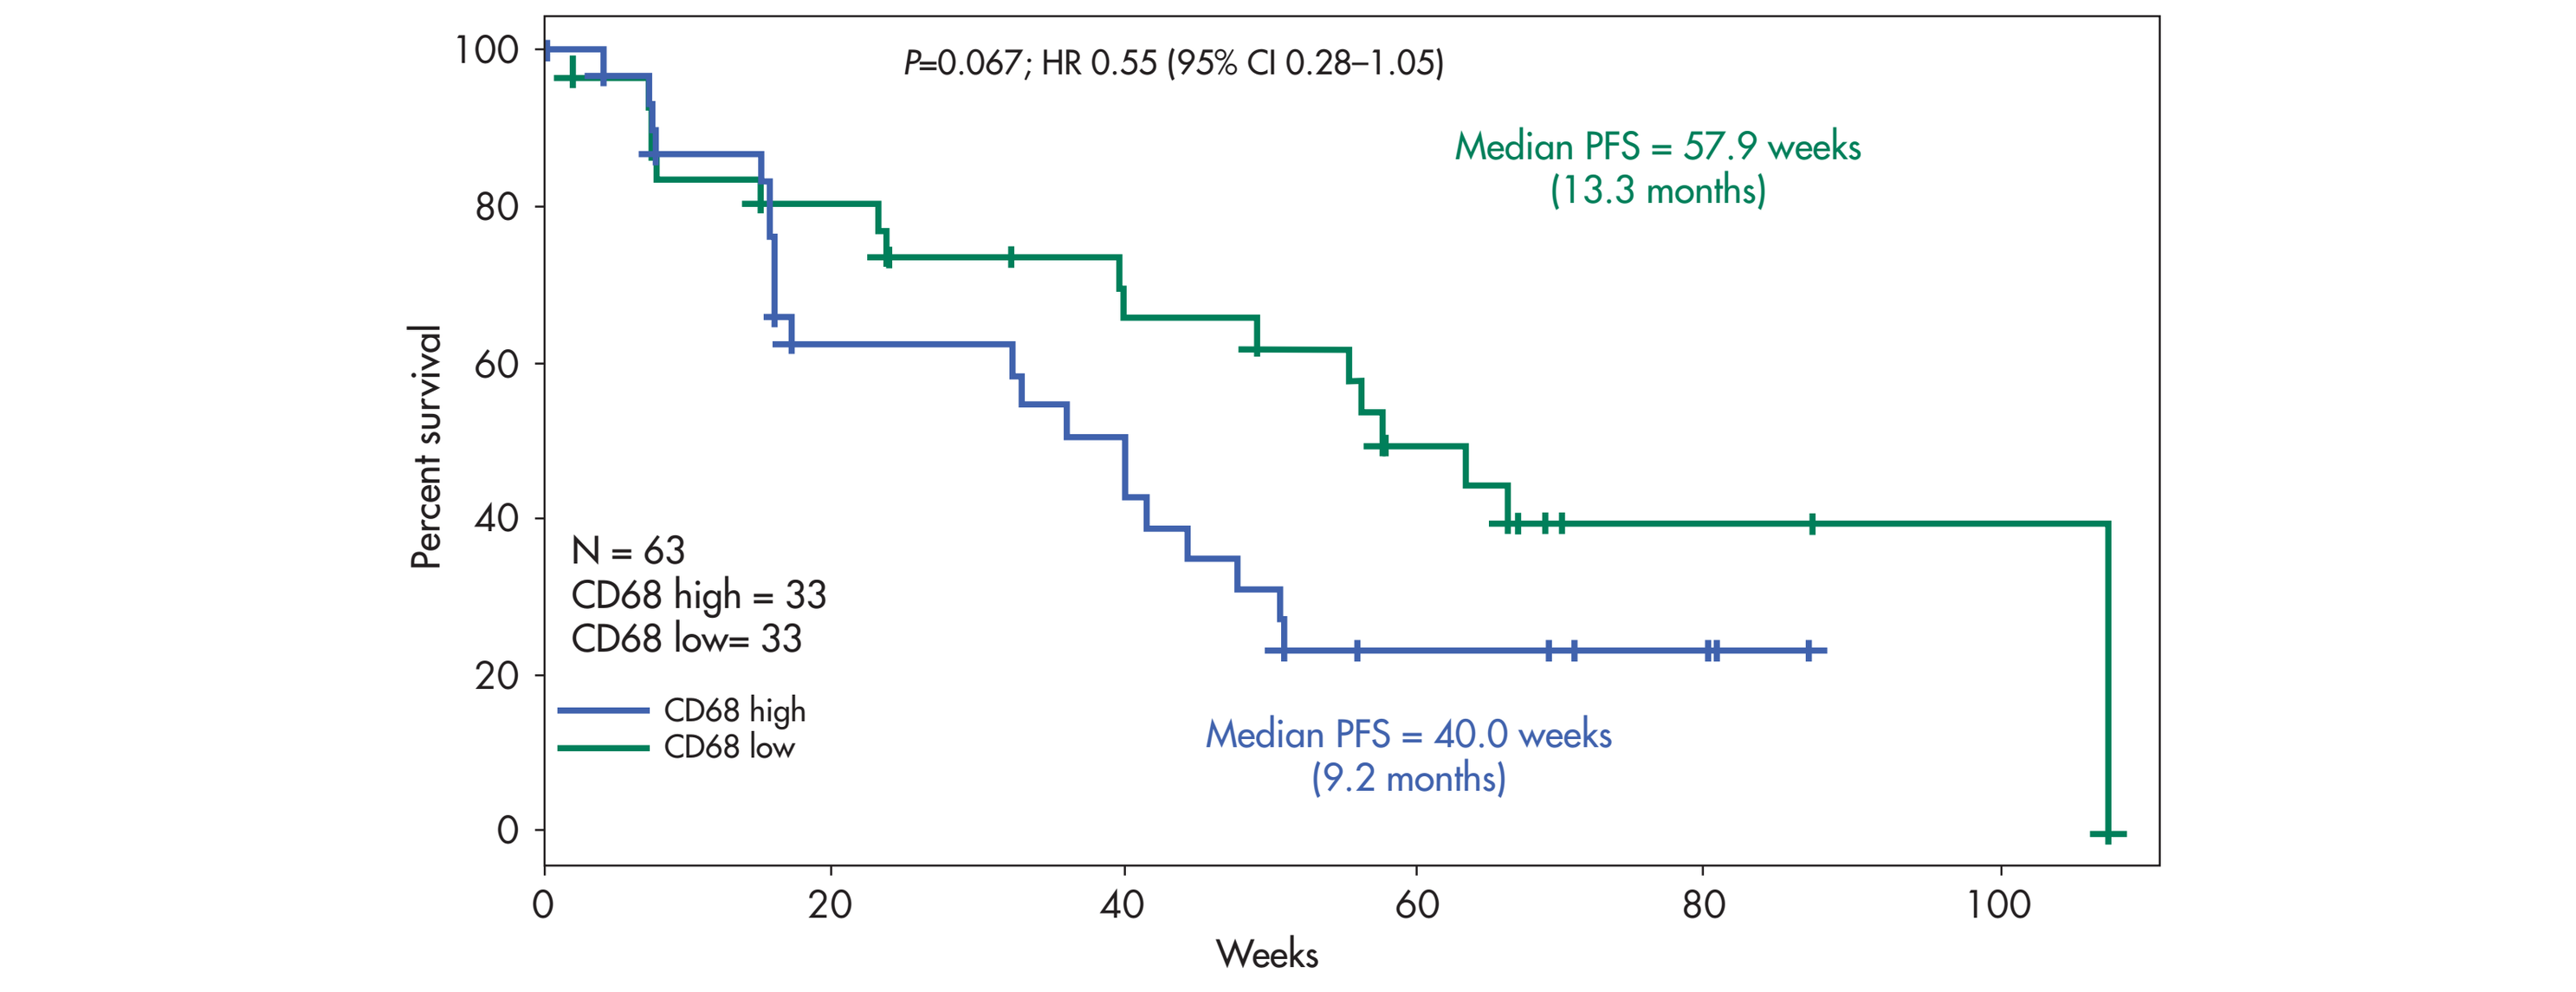
P=0.067; HR 0.55 (95% CI 0.28–1.05)
Median PFS = 57.9 weeks (13.3 months)
Median PFS = 40.0 weeks (9.2 months)
N = 63
CD68 high = 33
CD68 low= 33
CD68 high
CD68 low
Percent survival
Weeks

PFS, progression-free survival.

Figure 7. Example CD68 IHC From Human RCC.

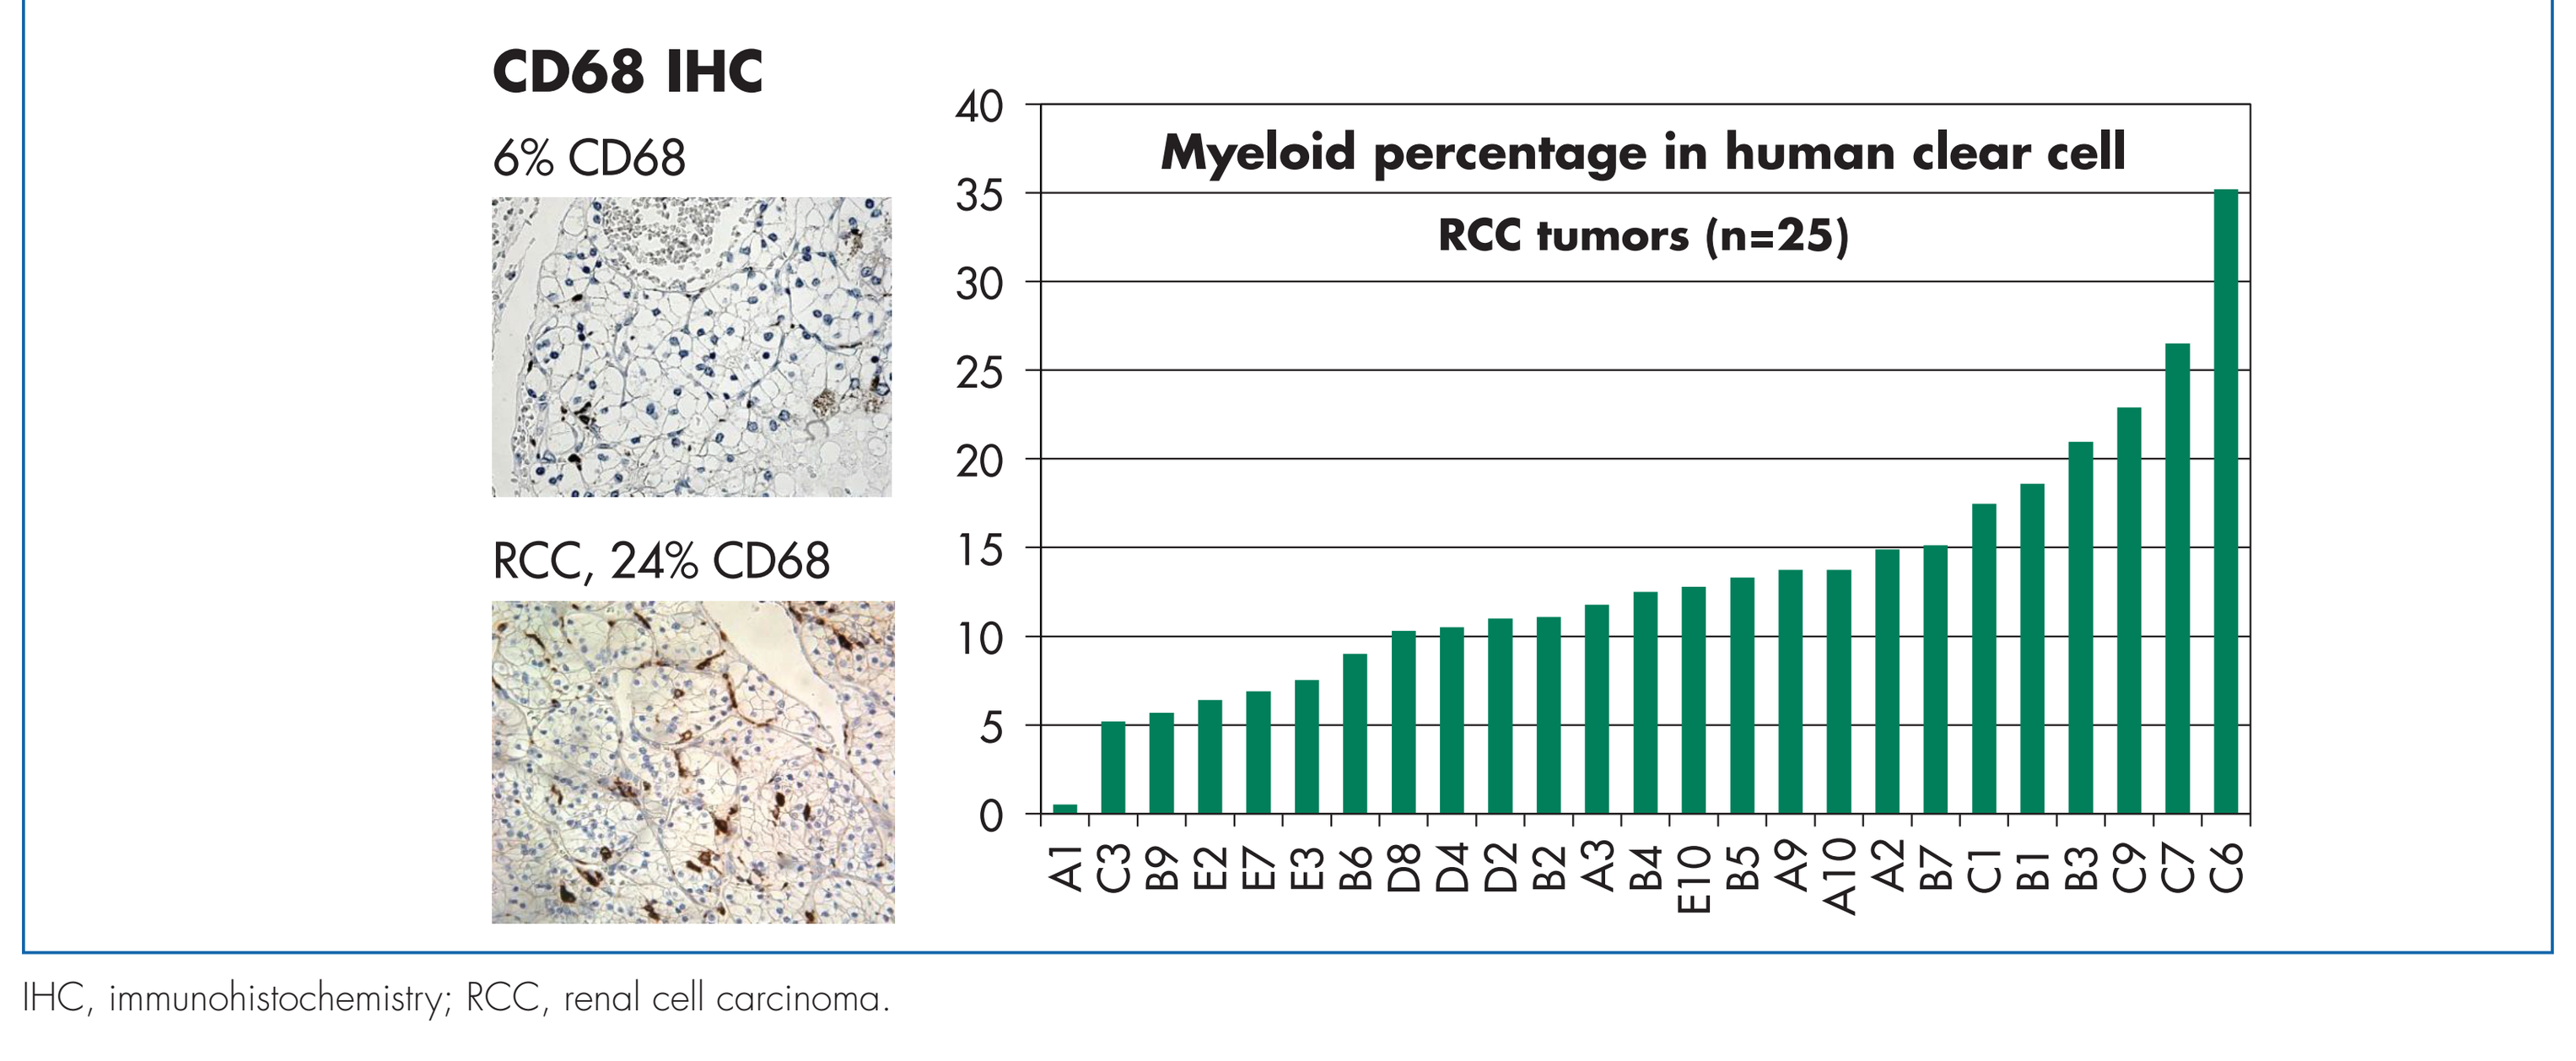
CD68 IHC
6% CD68
RCC, 24% CD68
Myeloid percentage in human clear cell RCC tumors (n=25)
A1 C3 B9 E2 E7 E3 B6 D8 D4 D2 B2 A3 B4 E10 B5 A9 A10 A2 B7 C1 B1 B3 C9 C7 C6

IHC, immunohistochemistry; RCC, renal cell carcinoma.

Conclusions

- A 24-gene RNA biomarker that defines a specific population of tumor-infiltrating myeloid cells identified a tivozanib-treated ccRCC population with significantly improved PFS
- This observation suggests the presence of a VEGF pathway–resistance mechanism associated with this myeloid population
- These results warrant the consideration of combination therapies targeting both the VEGF pathway and myeloid cells

References

1 Sonpavde G and Choueiri TK. *Urol Oncol*. 2014;32:5–15.
2 Watters JW, et al. *Cancer Res*. 2009;69:8949–8957.
3 Nosov DA, et al. *J Clin Oncol*. 2012;30:1678–1685.
4 Motzer RJ, et al. *J Clin Oncol*. 2013;31:3791–3799.
5 Hutson TE, et al. *J Clin Oncol*. 2012;30:Abstract TPS4686.
6 Zhao H, et al. *PLoS Med*. 2006;3:e13.

Acknowledgments

This study was supported by AVEO Oncology and Astellas. Editorial assistance was provided by Melissa Kirk, PhD, of Scientific Connexions, and was funded by AVEO Oncology and Astellas.

PRESENTED AT THE EUROPEAN SOCIETY FOR MEDICAL ONCOLOGY ANNUAL CONGRESS; SEPTEMBER 26–30, 2014; MADRID, SPAIN

Corresponding author: Bin Feng, bfeng@aveooncology.com. Poster will be available on-line at http://investor.aveooncology.com.